Deciphering synaptic defect in psychiatric disorders using machine learning for drug screening applications.

Loic Cousin[1], Sandra Horschitz[2], Aurore Vuidel[1], Arnaud Ogier[1], Juyong Yoon[1], Peter Sommer[1],Laurent Brino[1], Philipp Koch[2 *], Karen Schmitt[1]

[1] Ksilink, Strasbourg, France
[2] Hector Institute for Translational Brain Research (HITBR), Central Institute of Mental Health, University of Heidelberg, Mannheim, Germany
*Corresponding authors: karen.schmitt@ksilink.com, philipp.koch@zi-Mannheim.de

DISC1 as a trigger to model psychiatric disorders in iPSC-derived neurons

Isogenic DISC1 disrupted lines *(Srikanth et al, Cell Reports, 2015)*

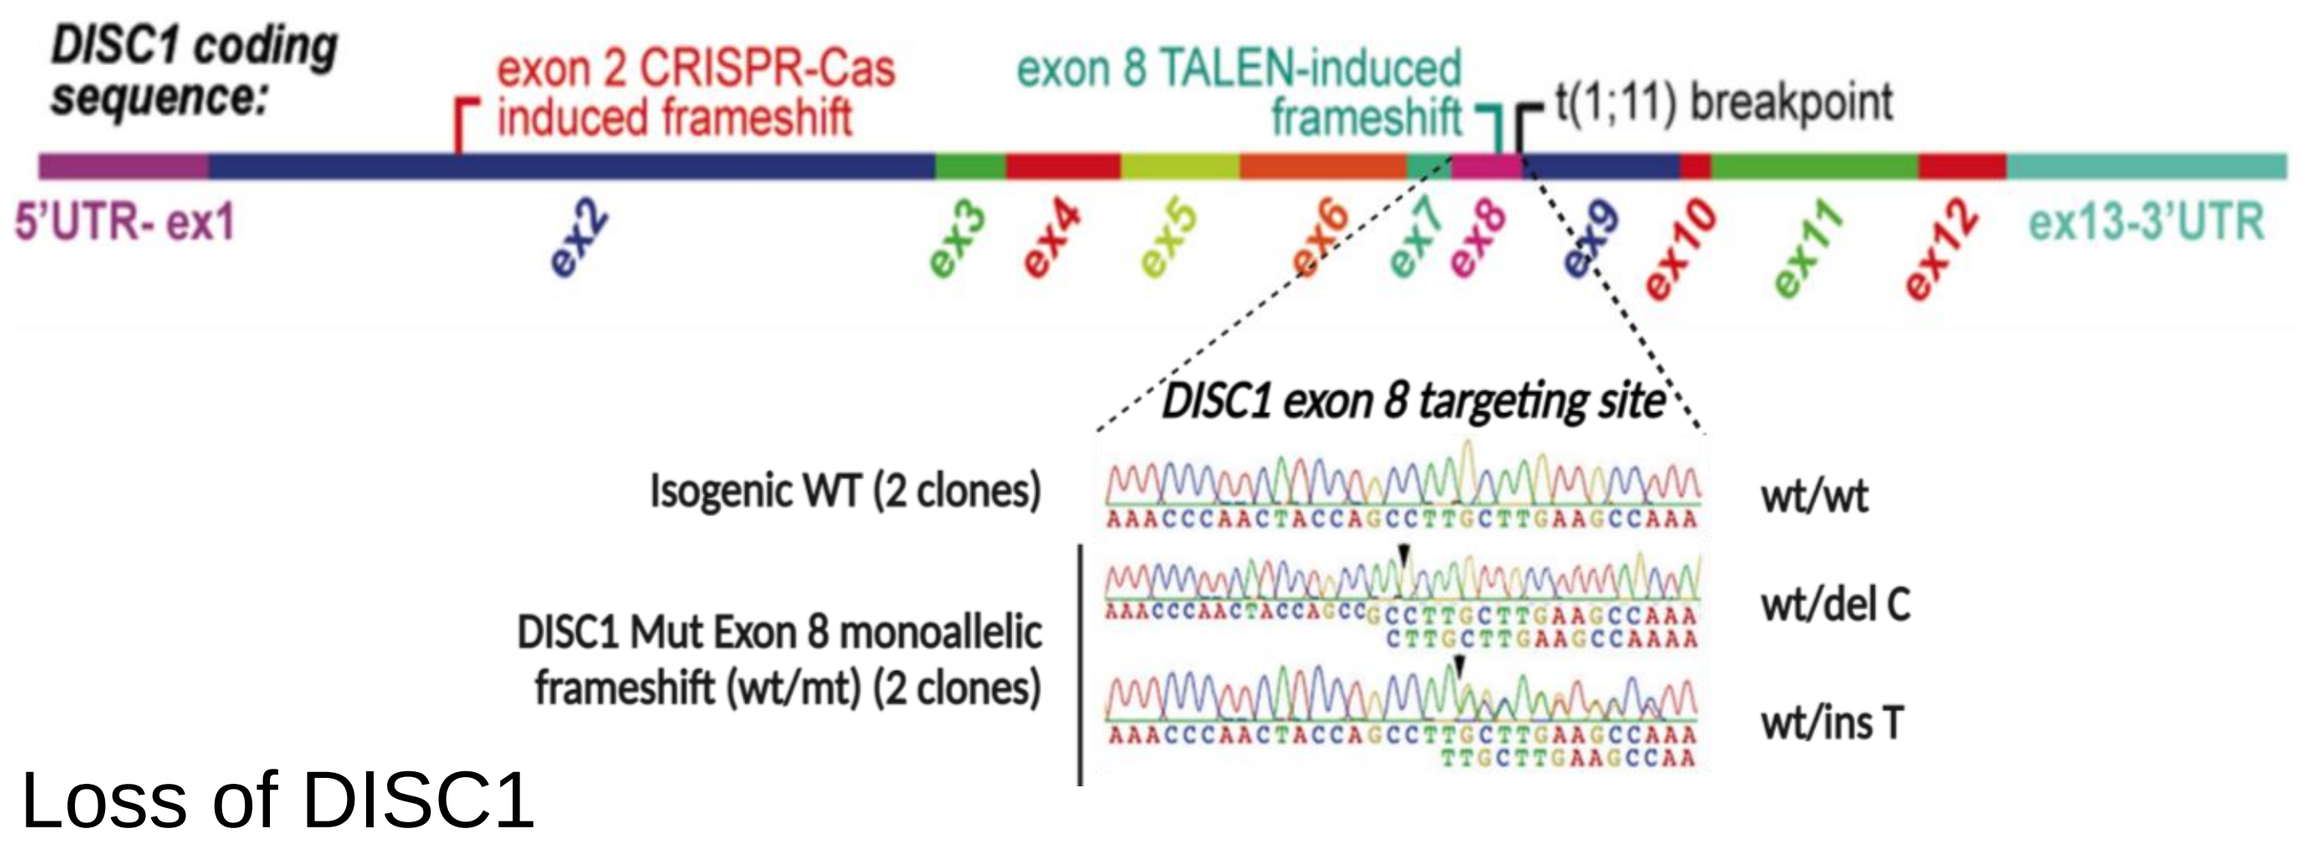

DISC1 coding sequence:
exon 2 CRISPR-Cas induced frameshift
exon 8 TALEN-induced frameshift
t(1;11) breakpoint
5'UTR- ex1
ex2
ex3
ex4
ex5
ex6
ex7
ex8
ex9
ex10
ex11
ex12
ex13-3'UTR
DISC1 exon 8 targeting site
Isogenic WT (2 clones)
DISC1 Mut Exon 8 monoallelic frameshift (wt/mt) (2 clones)
wt/wt
wt/del C
wt/ins T

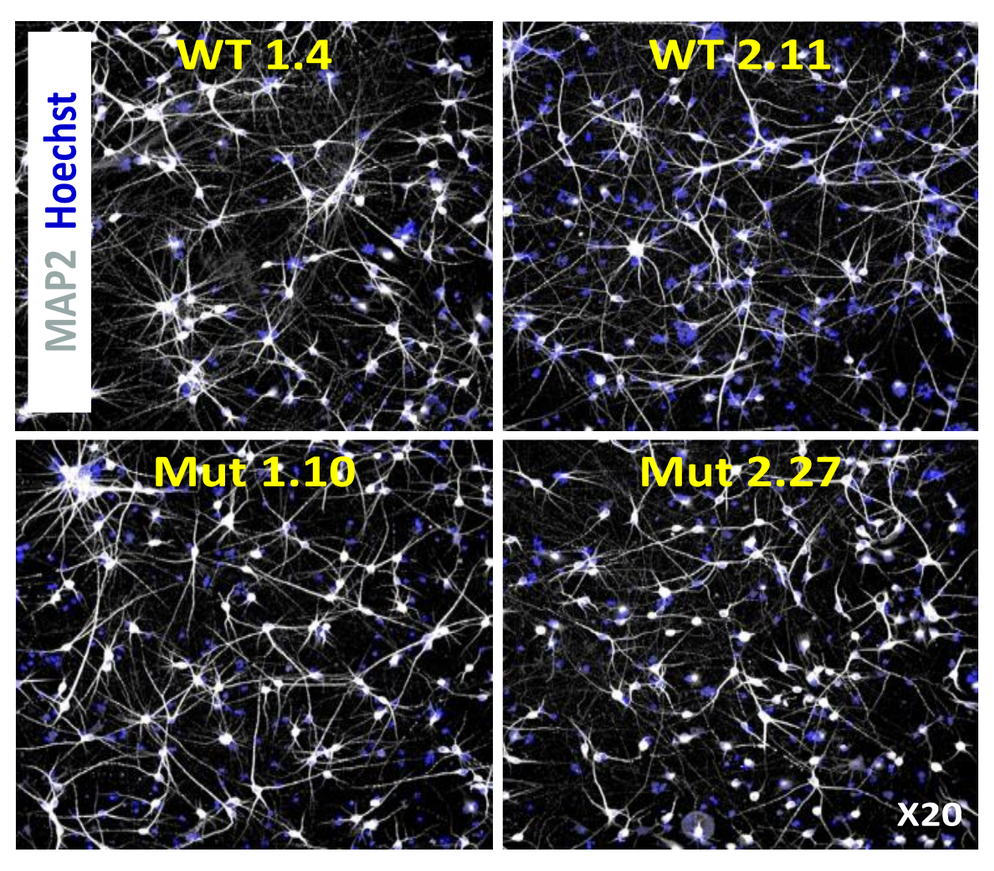

MAP2 Hoechst
WT 1.4
WT 2.11
Mut 1.10
Mut 2.27
X20

- Loss of DISC1
- Decreased expression of neuronal markers & neurite outgrowth
- Increased proliferation and activation of WNT signaling
- Decreased expression of synaptic proteins
- Impaired neuronal activity (Ca^{2+} imaging)

Automated workflow for multi-parametric phenotypic profiling

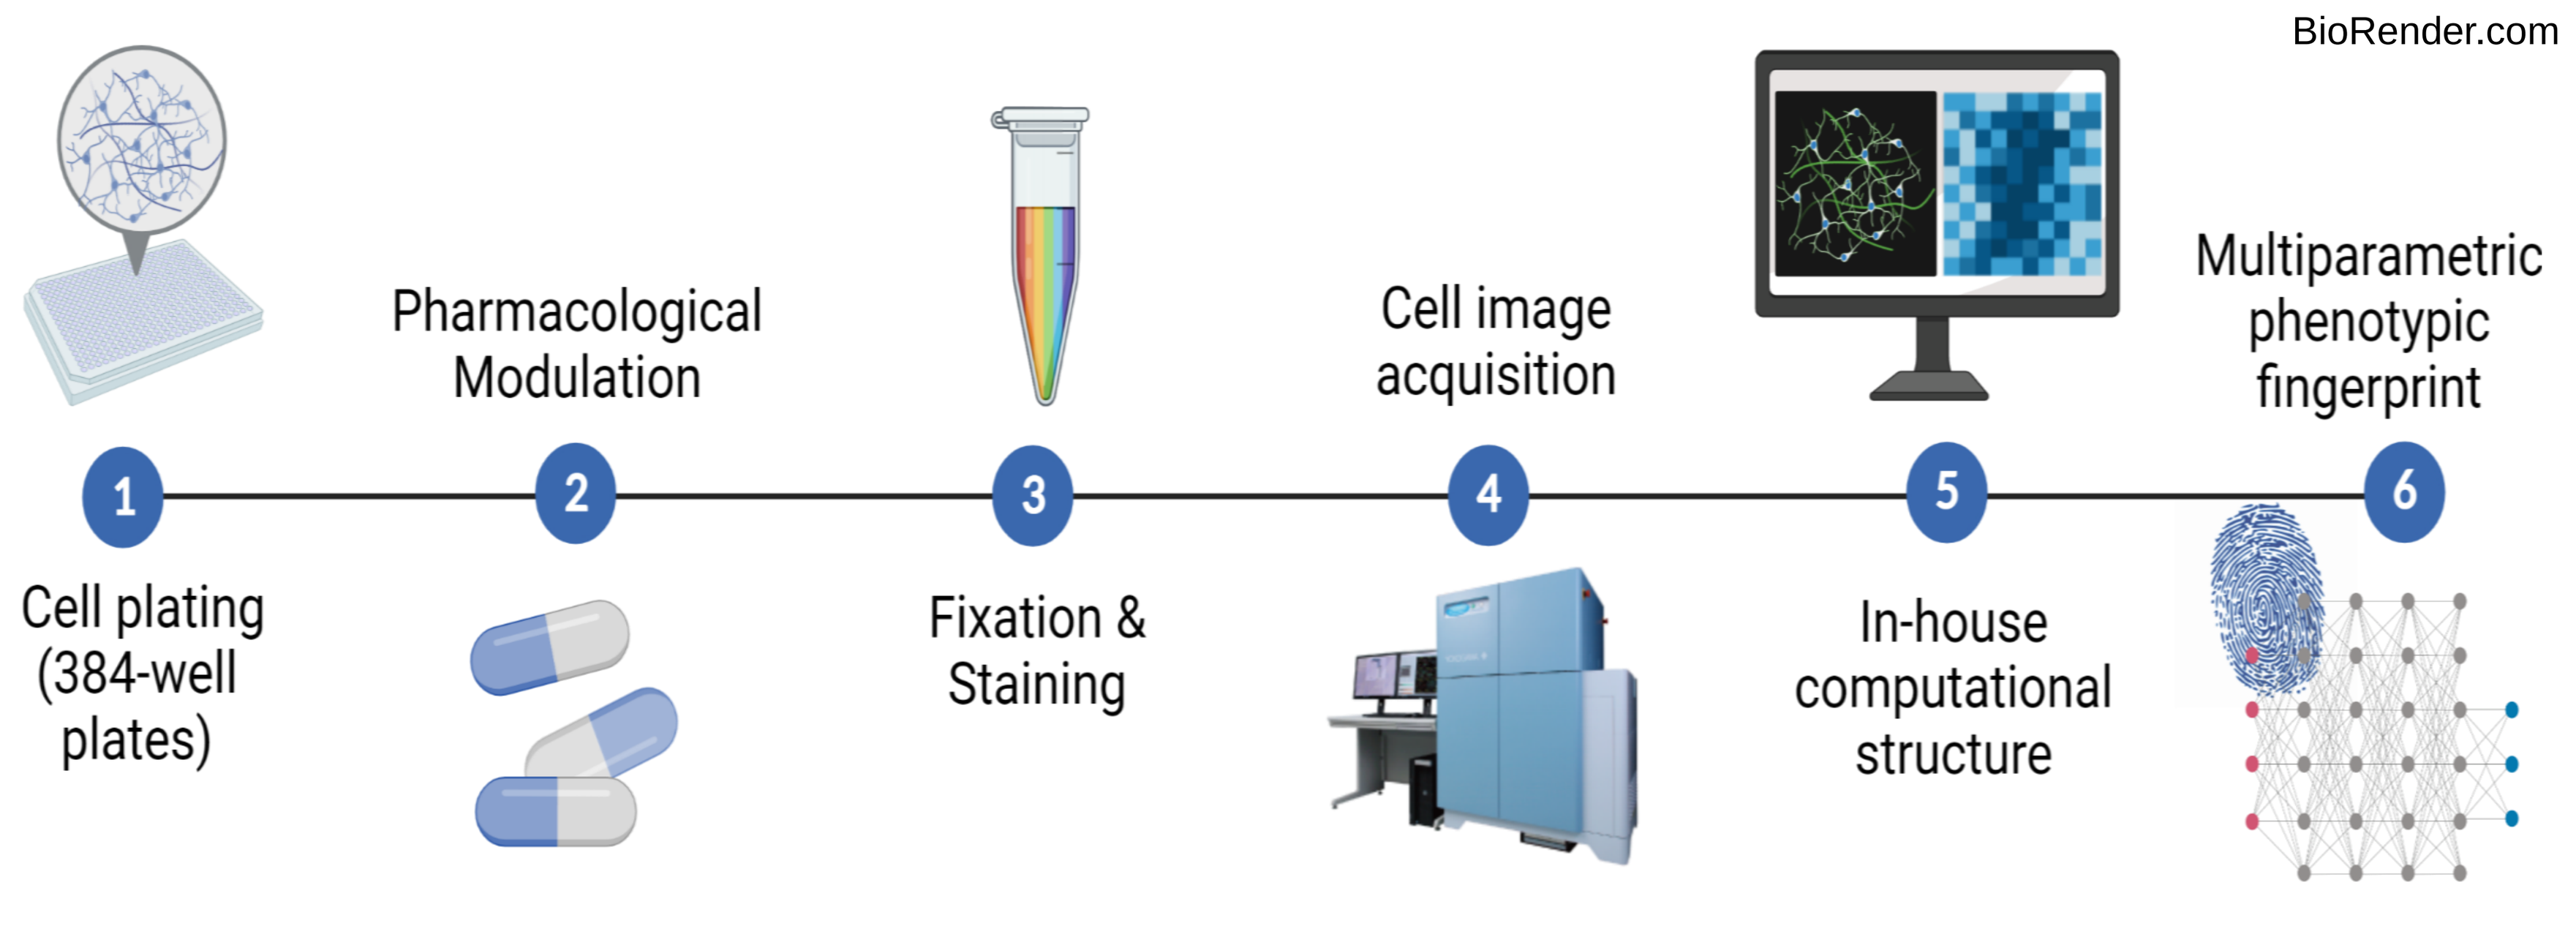

Created with BioRender.com
1 Cell plating (384-well plates)
2 Pharmacological Modulation
3 Fixation & Staining
4 Cell image acquisition
5 In-house computational structure
6 Multiparametric phenotypic fingerprint

DISC1- triggered developmental synaptopathologies in iPSC-derived neurons

Synaptic staining panel in isogenic iPSC-derived neurons

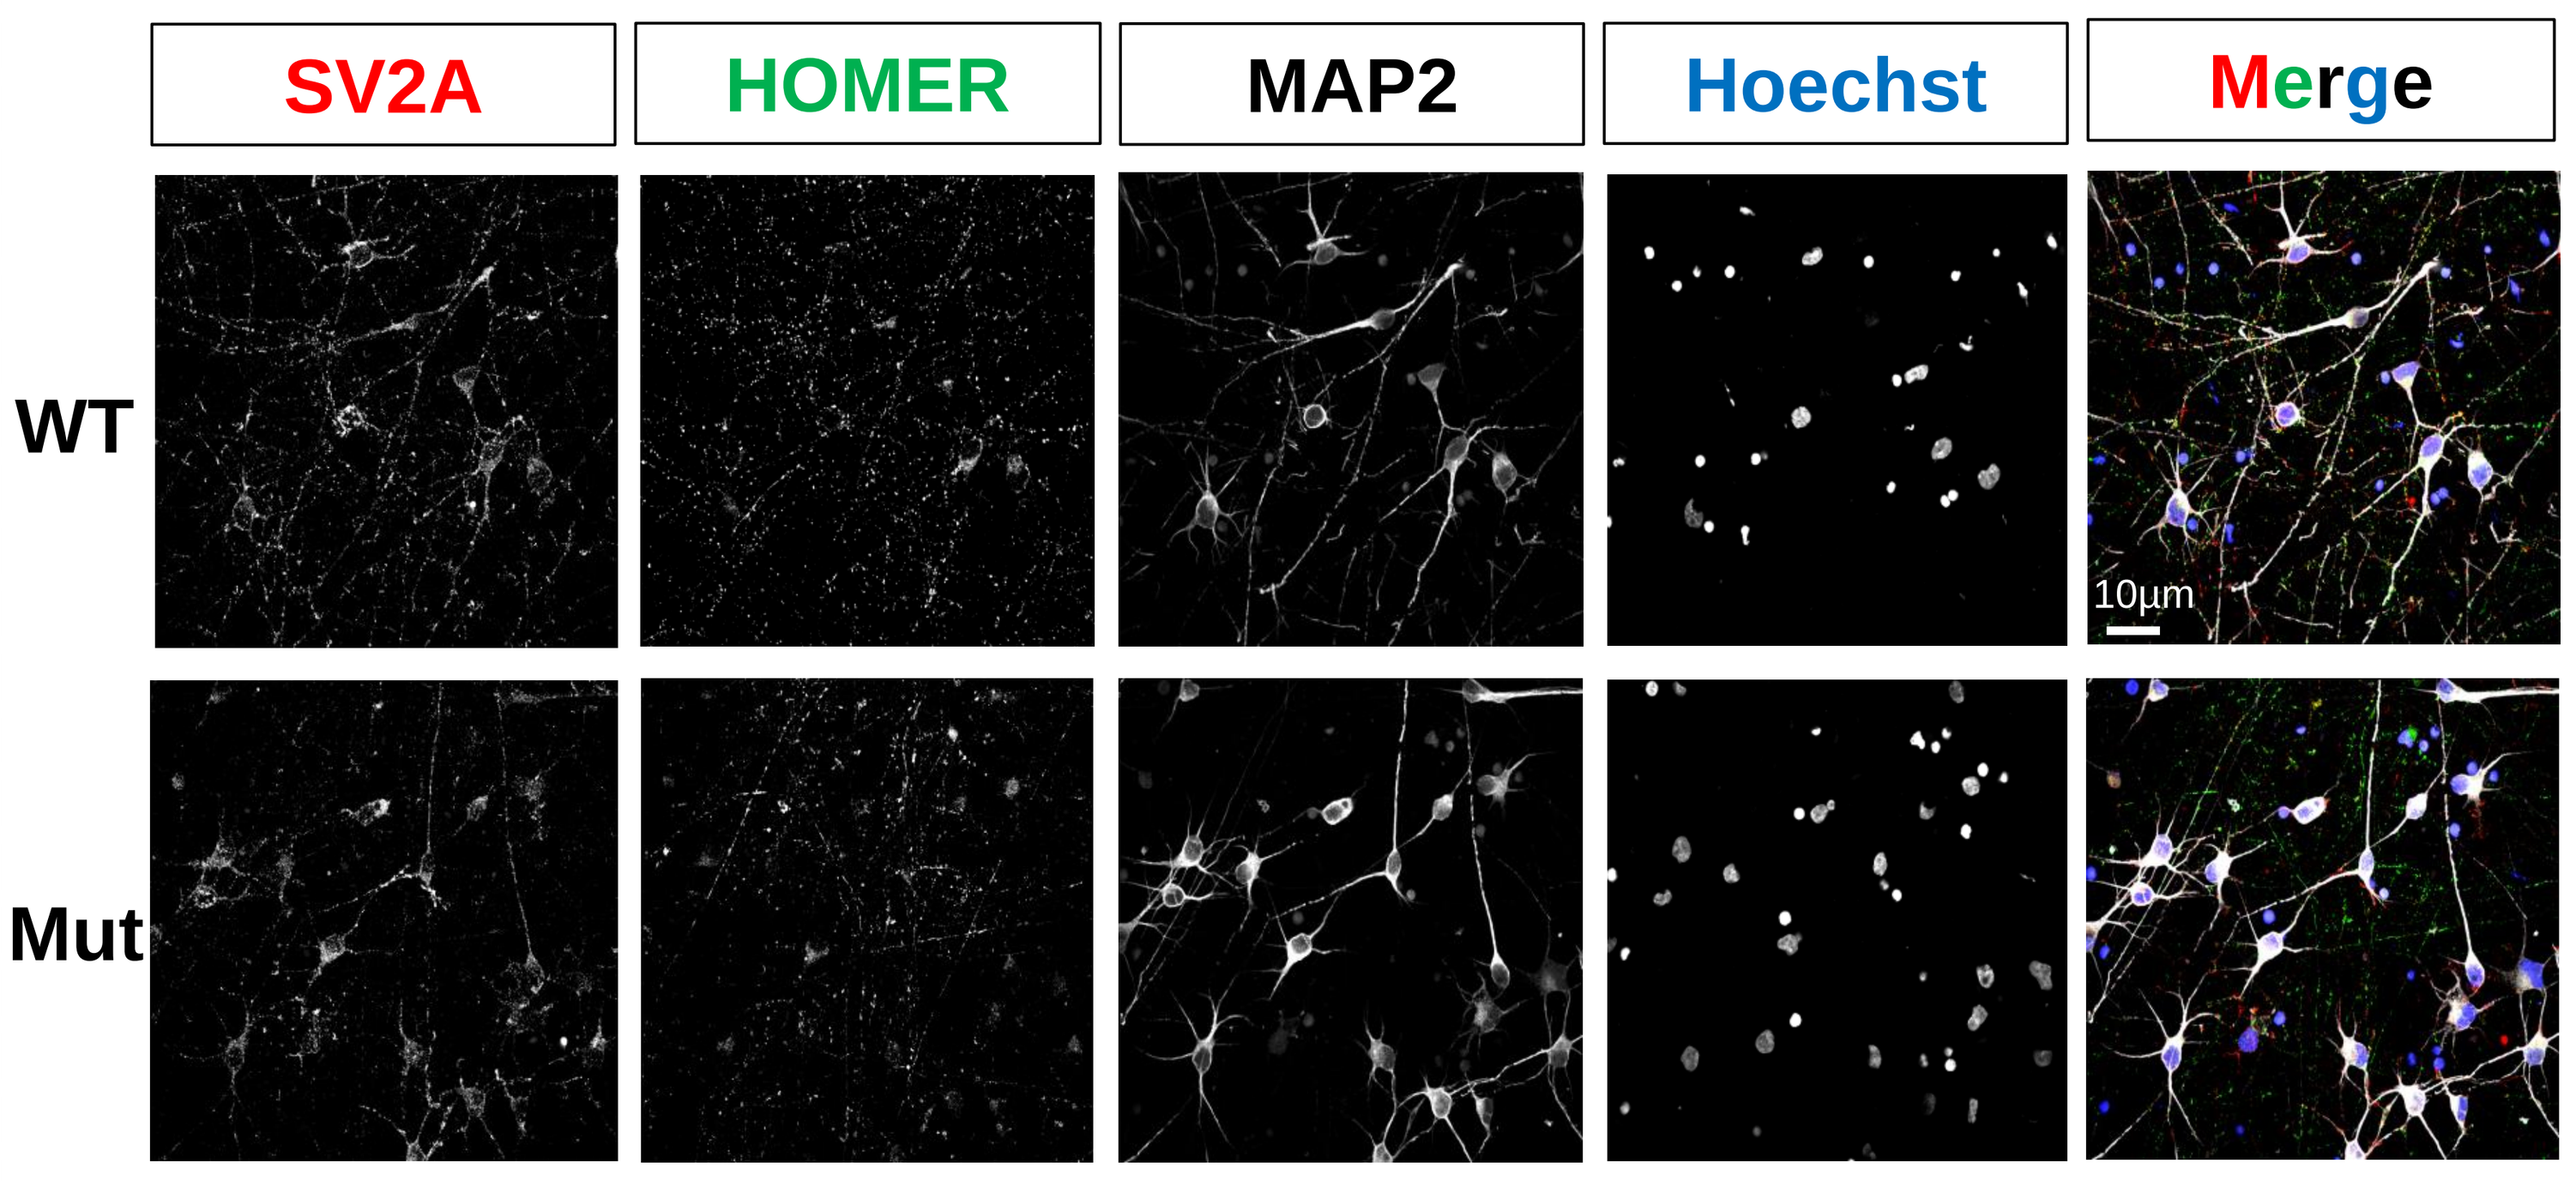

SV2A
HOMER
MAP2
Hoechst
Merge
WT
Mut
10µm

Feature extraction

Segmentation of Map2 (network complexity)

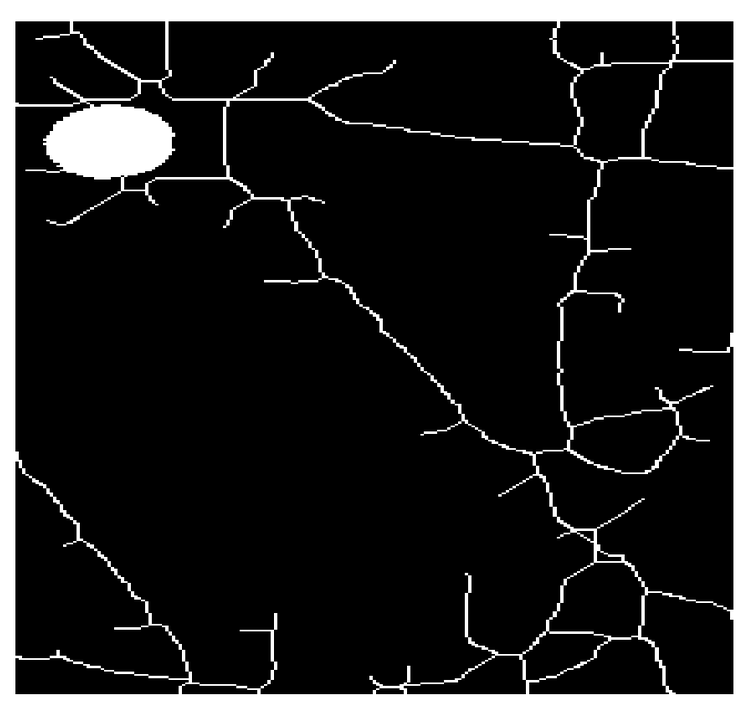

Detection of synaptic markers

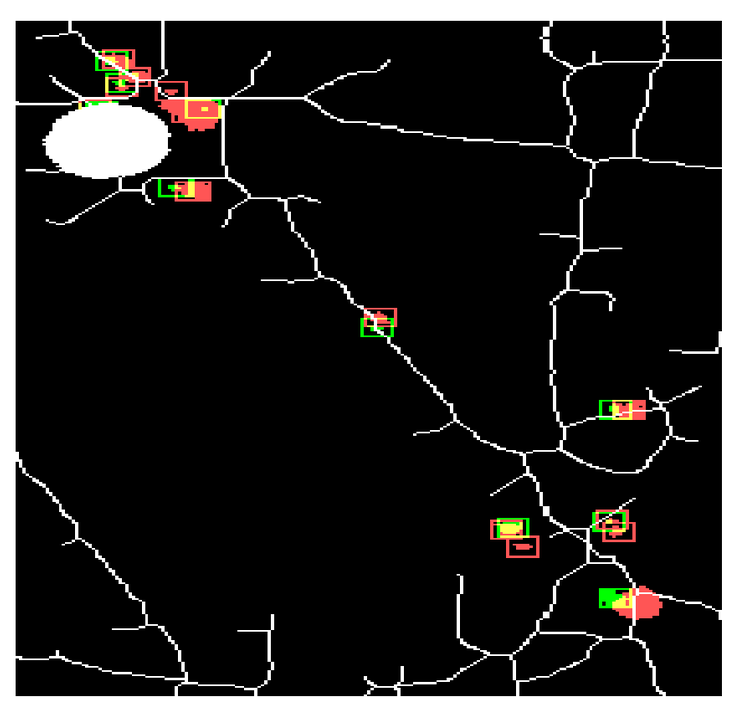

Examples of single extracted neuronal features

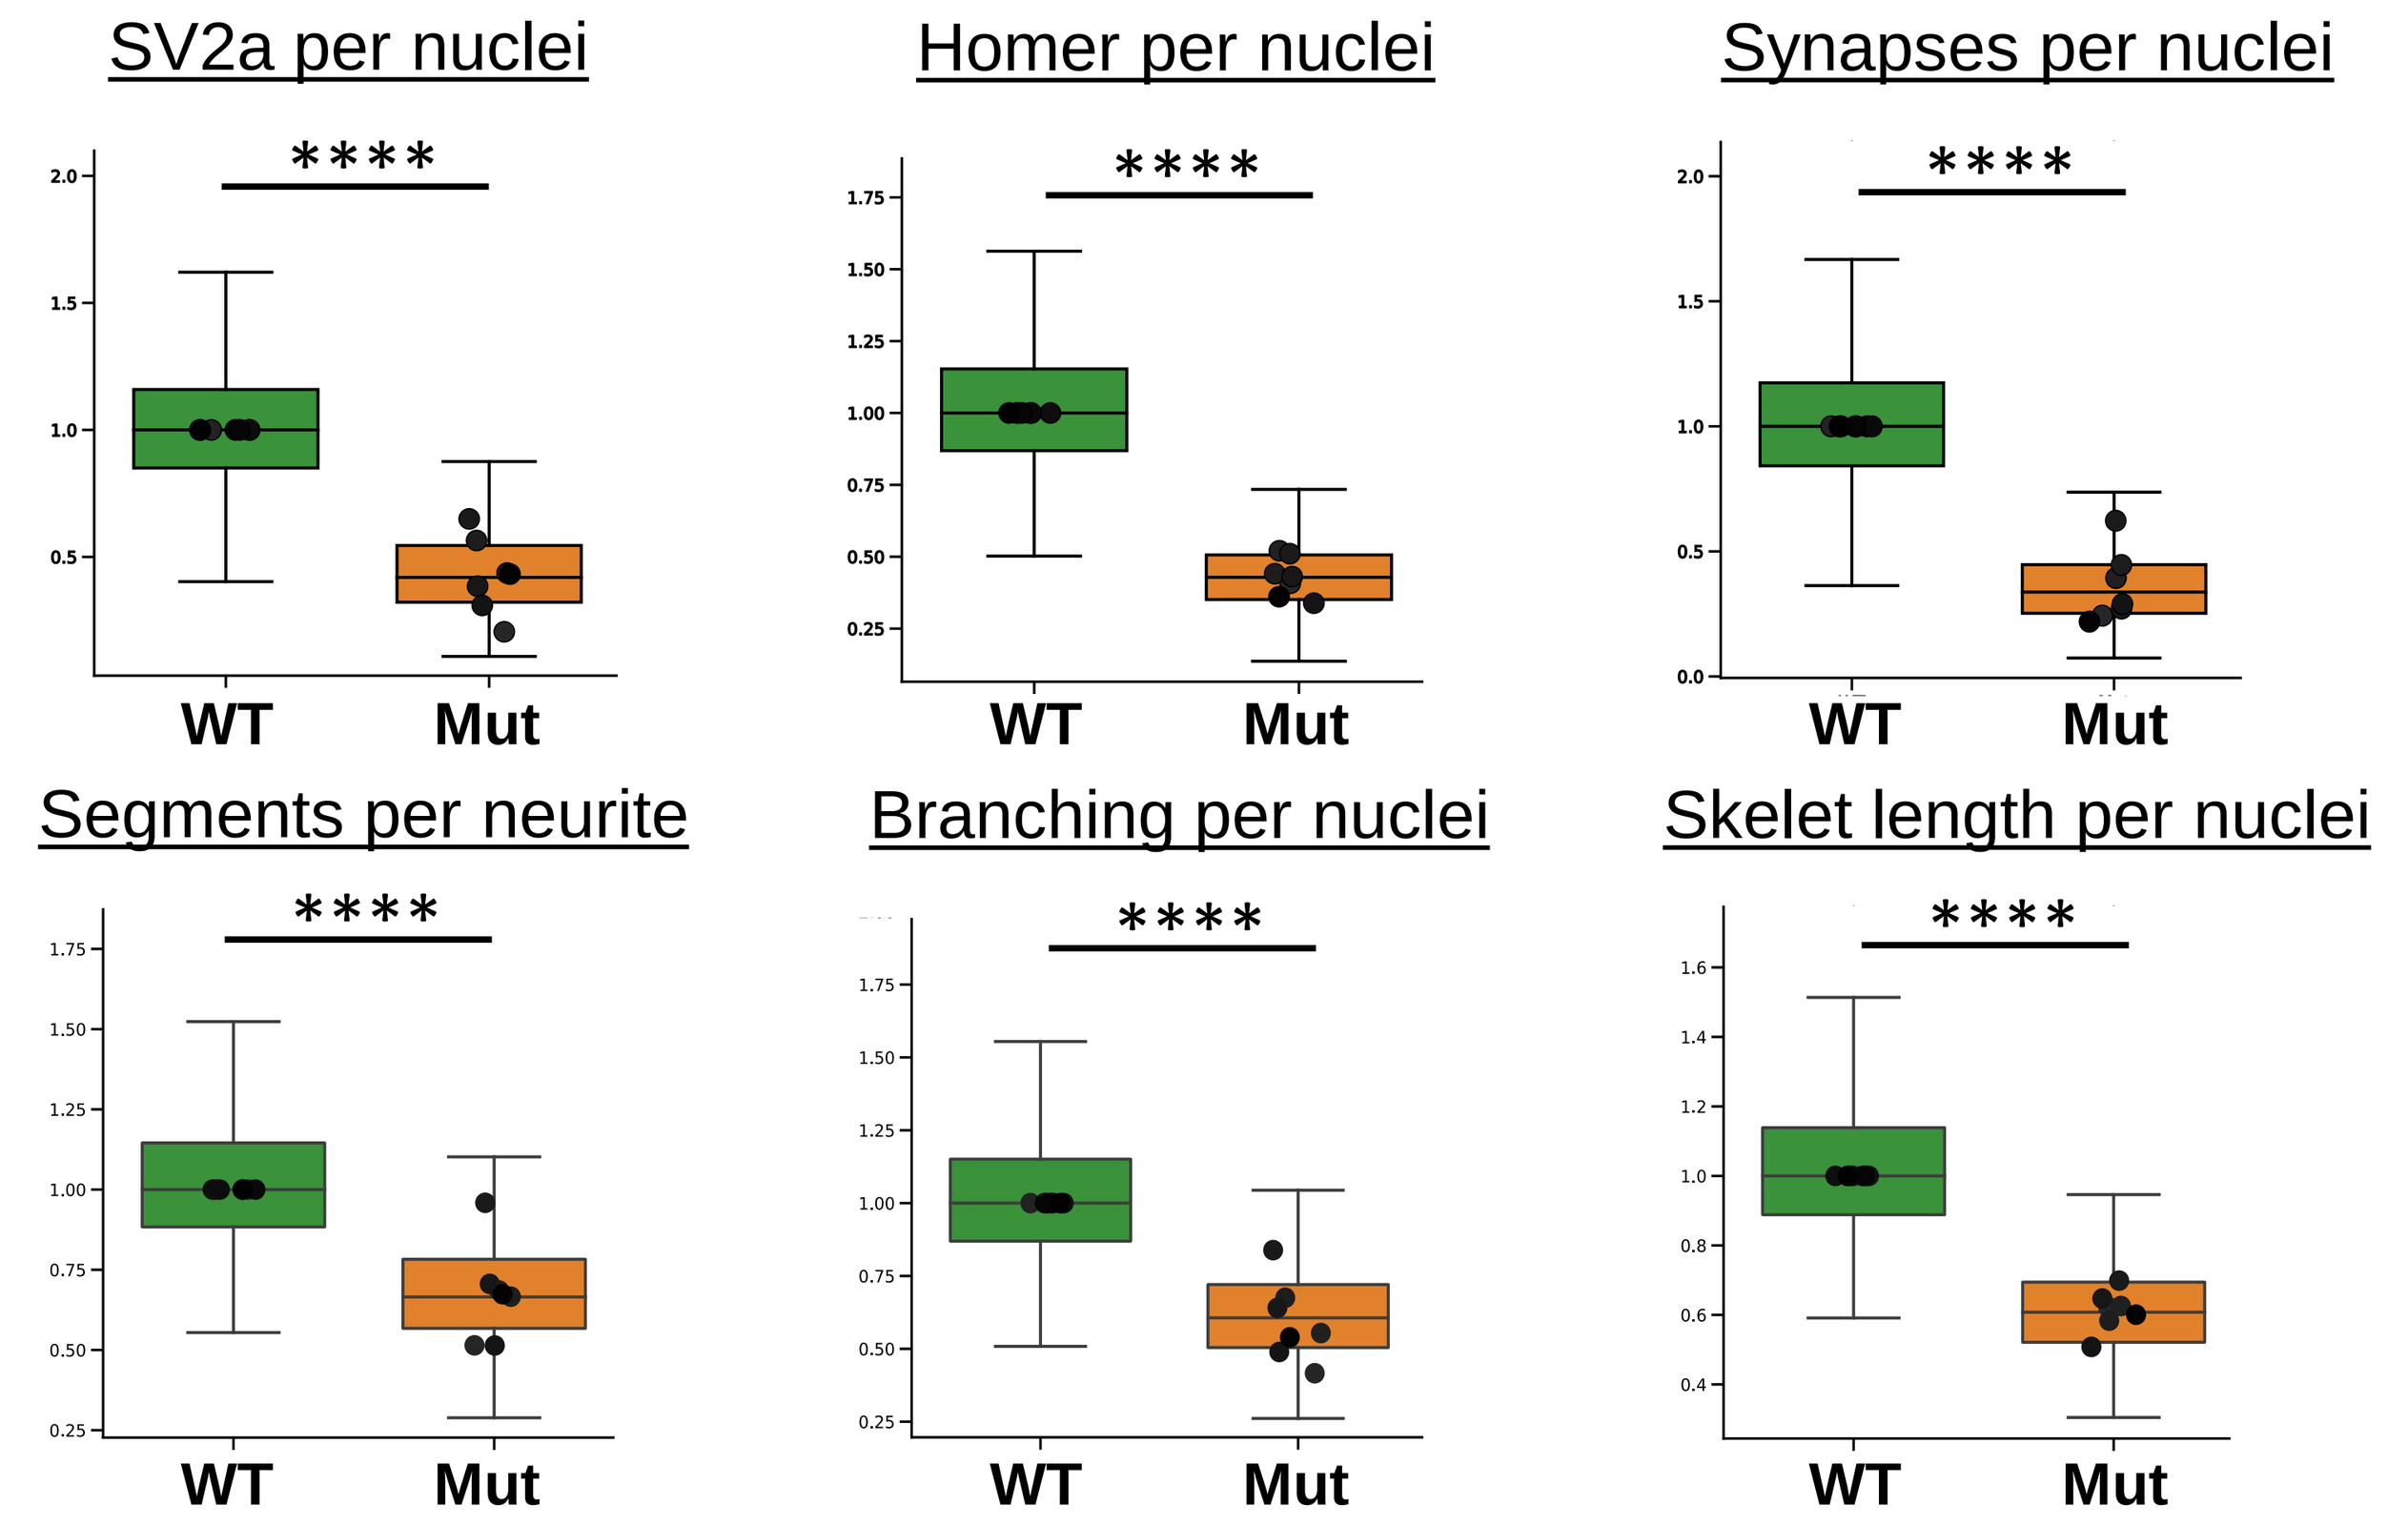

SV2a per nuclei
Homer per nuclei
Synapses per nuclei
Segments per neurite
Branching per nuclei
Skelet length per nuclei

WT
Mut

Machine Learning (ML) strategy to classify neurons based on multiple image-derived features

"Unseen" conditions such as novel cell lines or tested chemical compounds can be mapped relative to the two reference classes.

Computational workflow

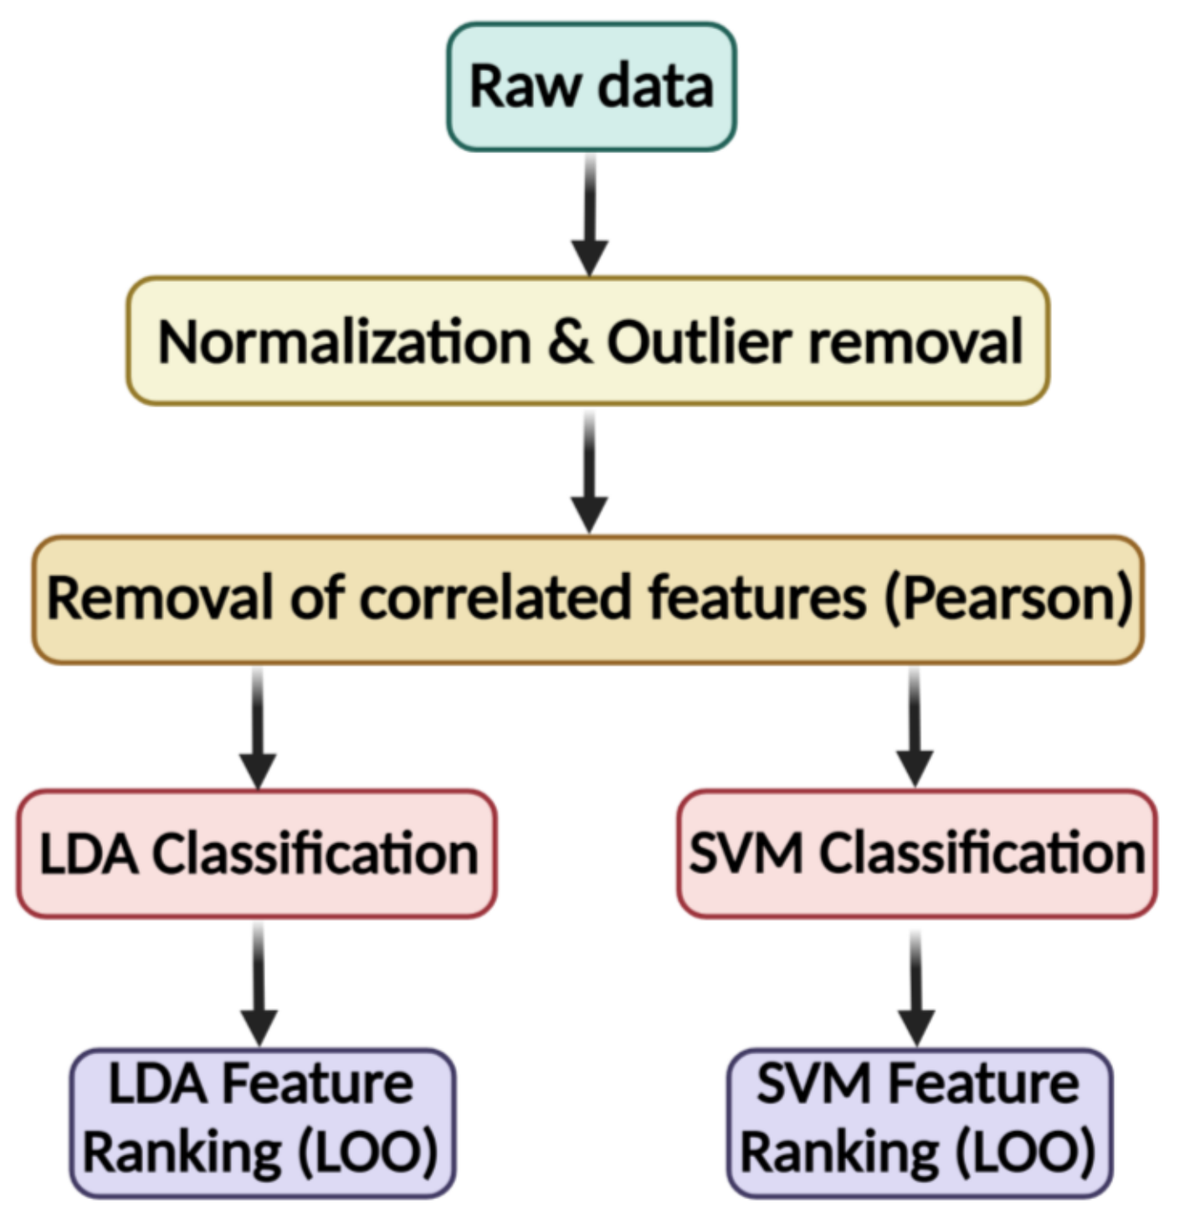

Raw data
Normalization & Outlier removal
Removal of correlated features (Pearson)
LDA Classification
SVM Classification
LDA Feature Ranking (LOO)
SVM Feature Ranking (LOO)

1 Pearson's correlation of the features

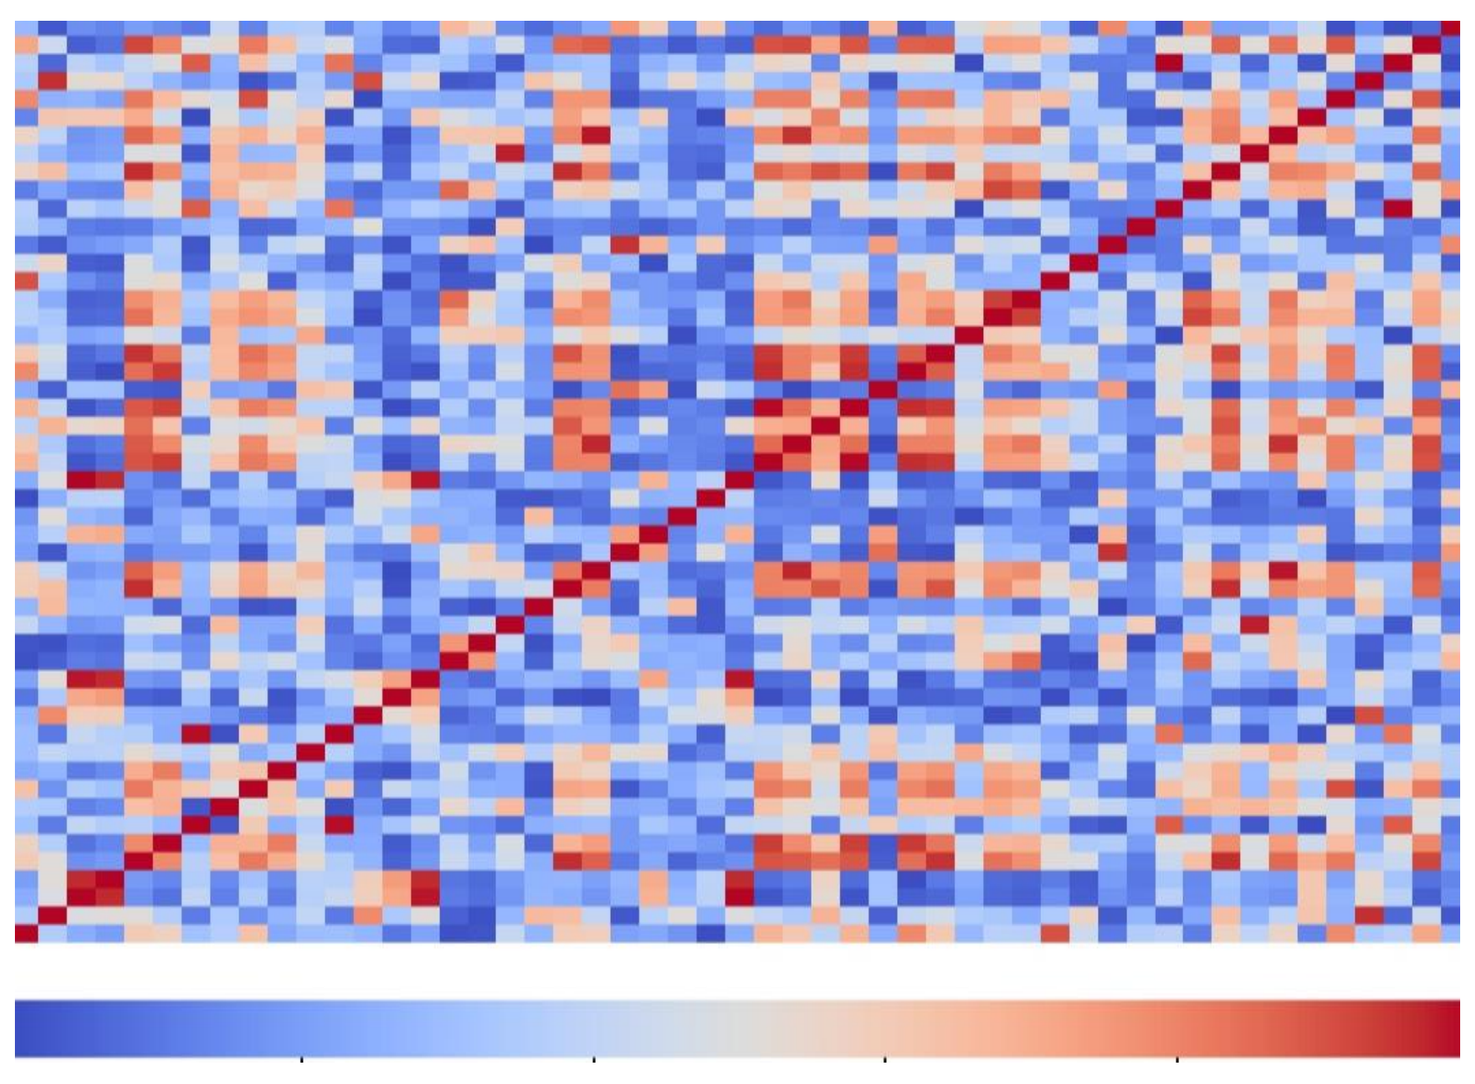

2 Machine learning of human iPSC-derived cortical neurons using Hoechst, SV2a, Homer, MAP2 staining

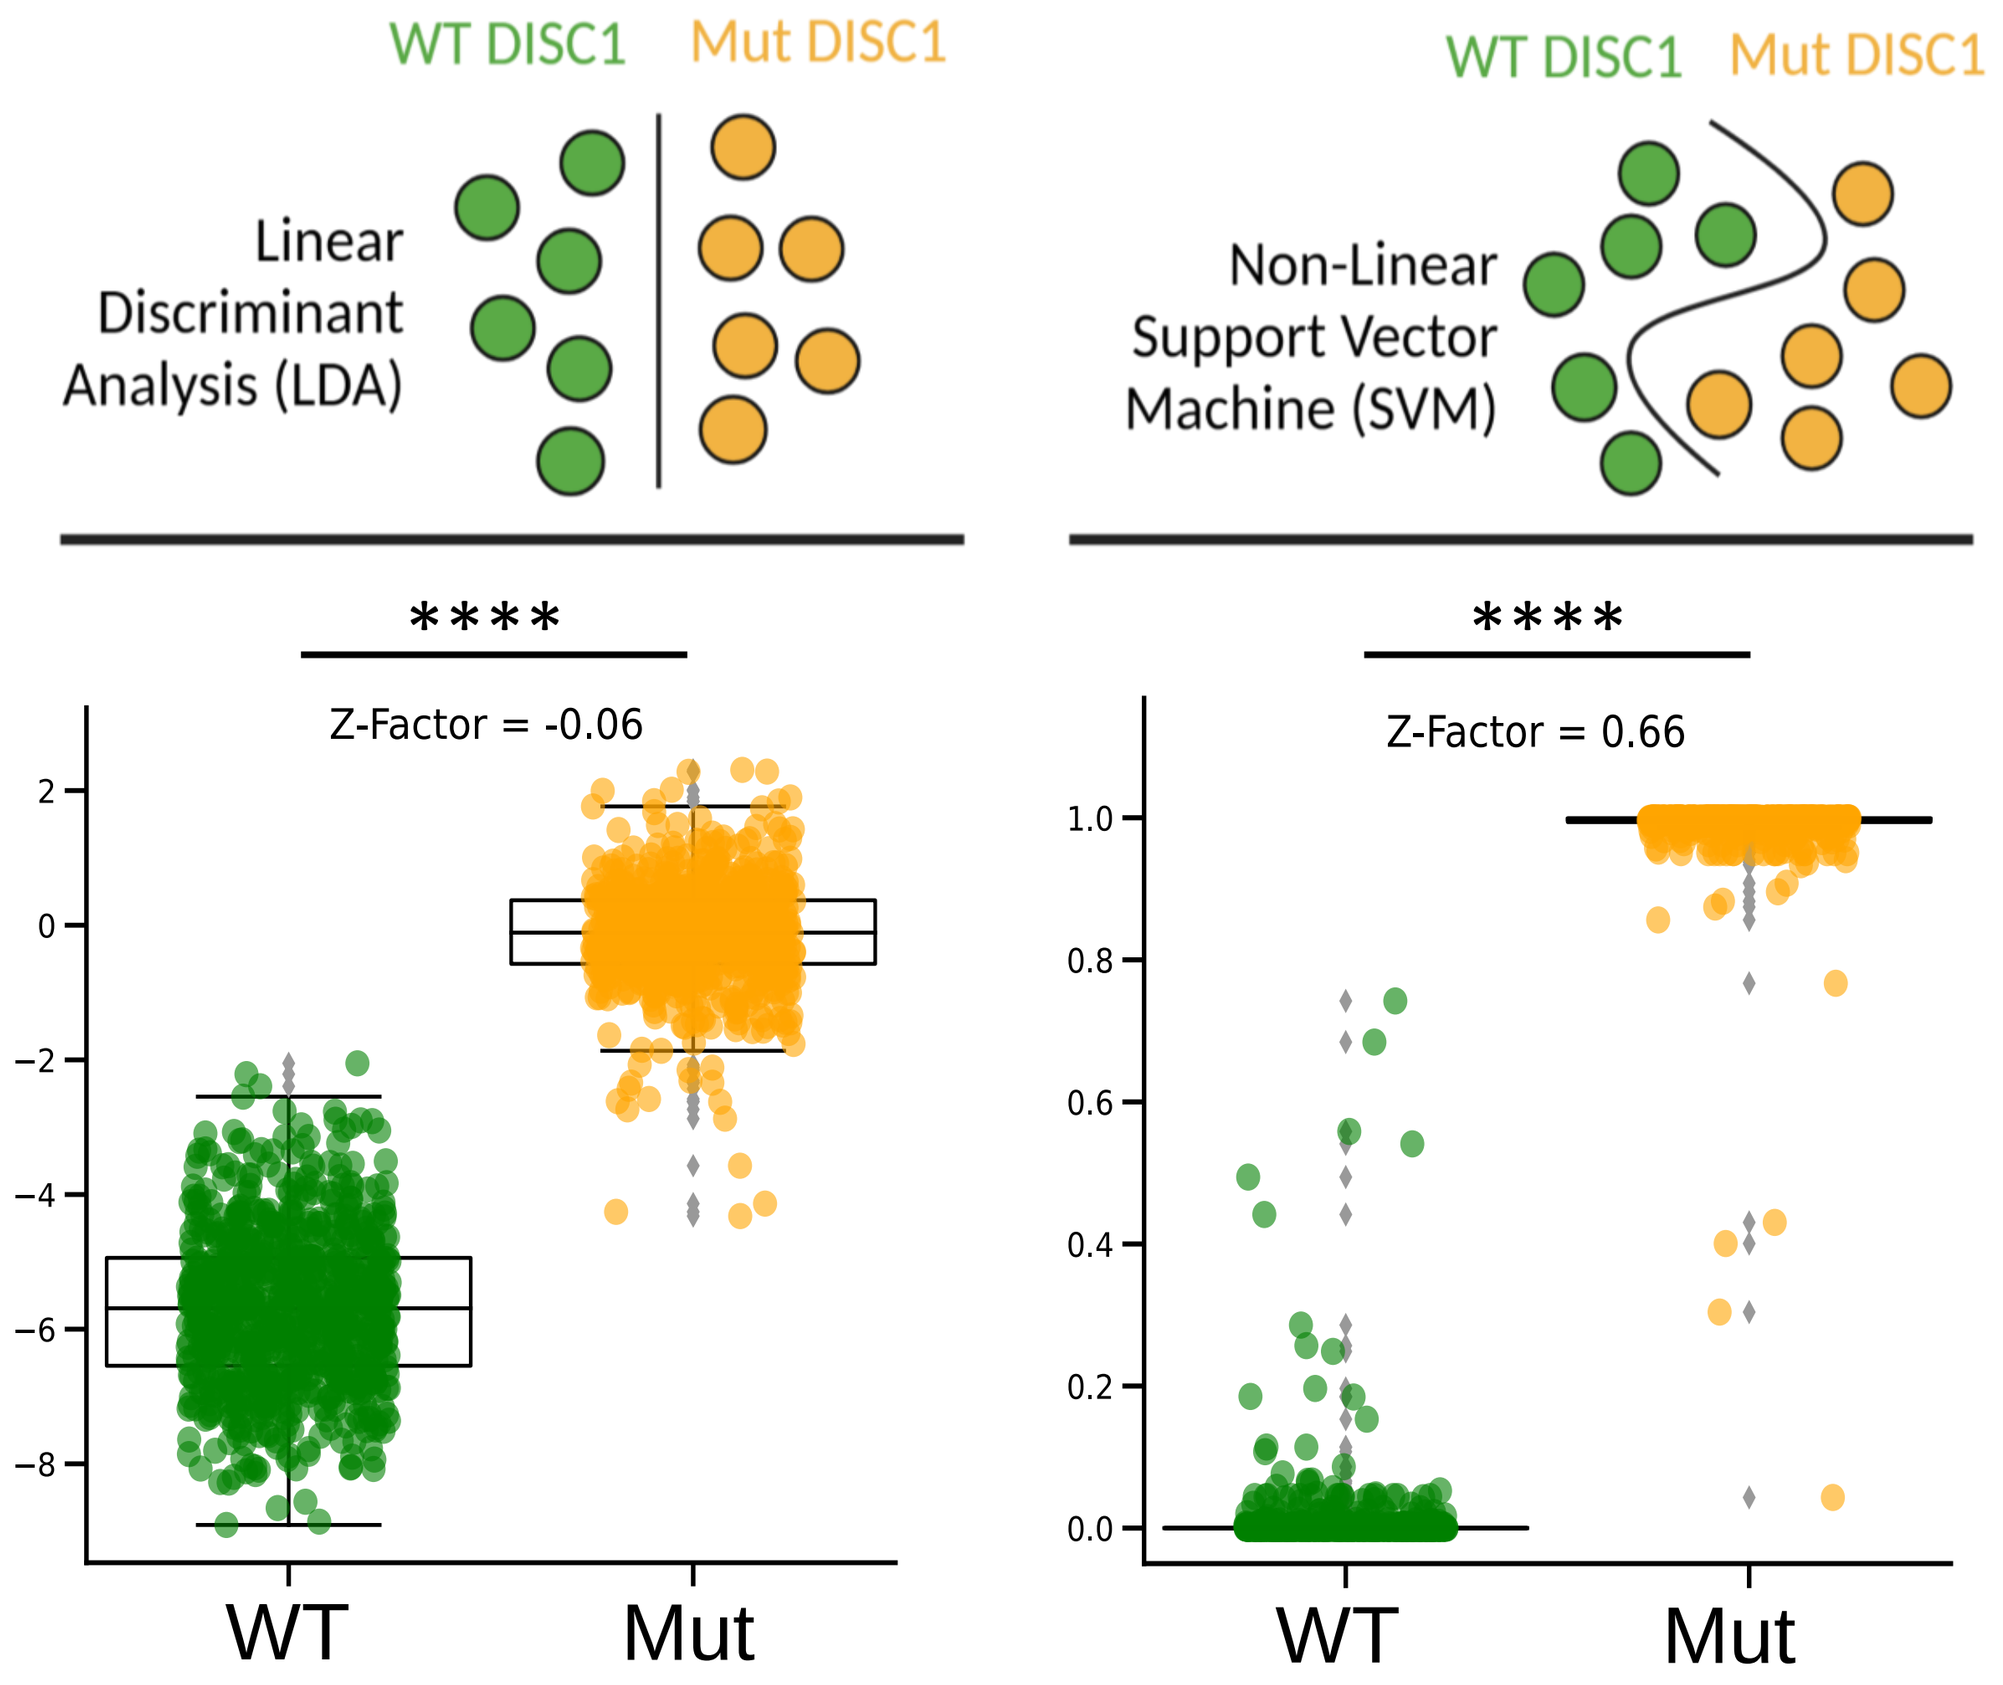

WT DISC1
Mut DISC1
Linear Discriminant Analysis (LDA)
Non-Linear Support Vector Machine (SVM)

Z-Factor = -0.06
Z-Factor = 0.66
WT
Mut

3 Features importance ranking

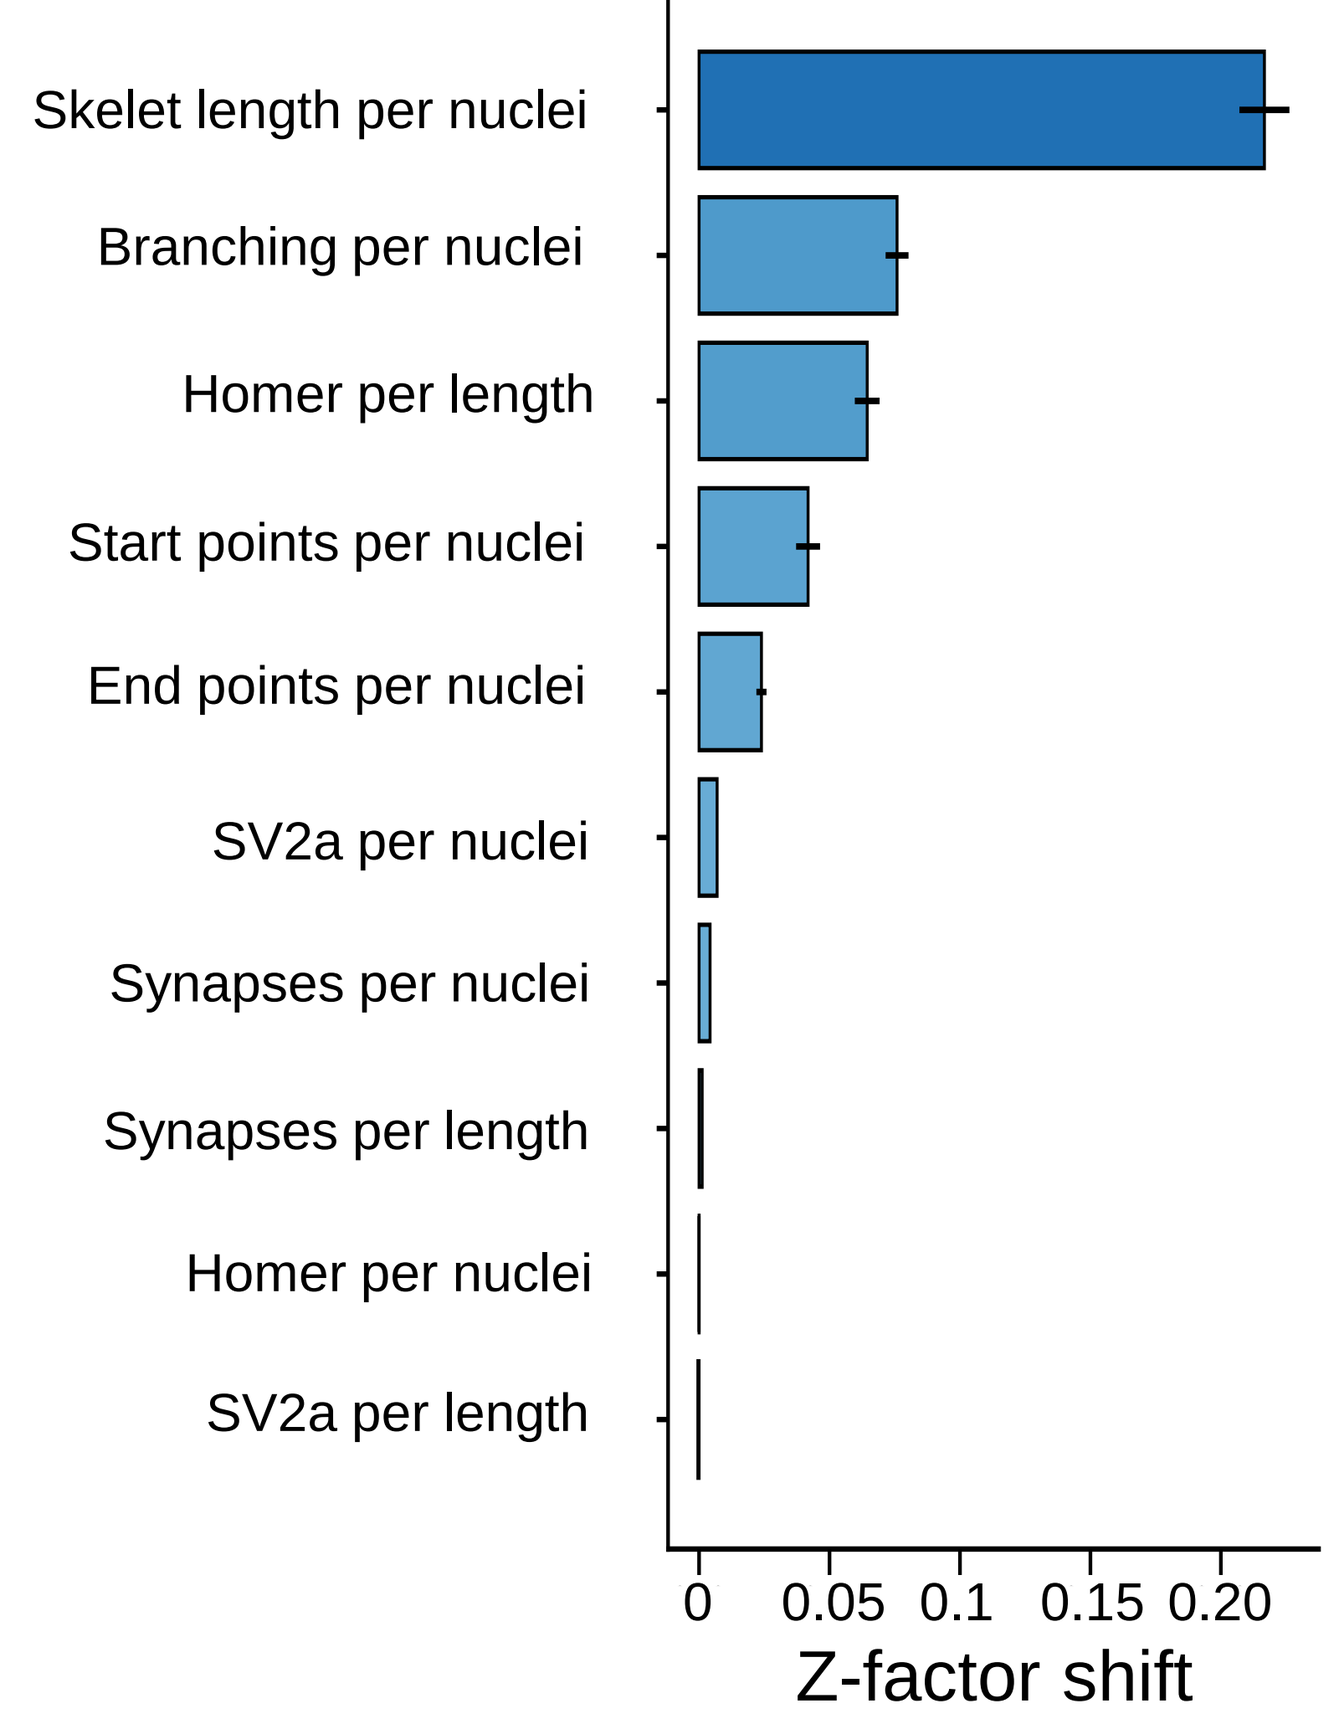

Skelet length per nuclei
Branching per nuclei
Homer per length
Start points per nuclei
End points per nuclei
SV2a per nuclei
Synapses per nuclei
Synapses per length
Homer per nuclei
SV2a per length
0
0.05
0.1
0.15
0.20
Z-factor shift

Classification of pharmacological modulation by multiparametric fingerprinting

Rolipram (R)

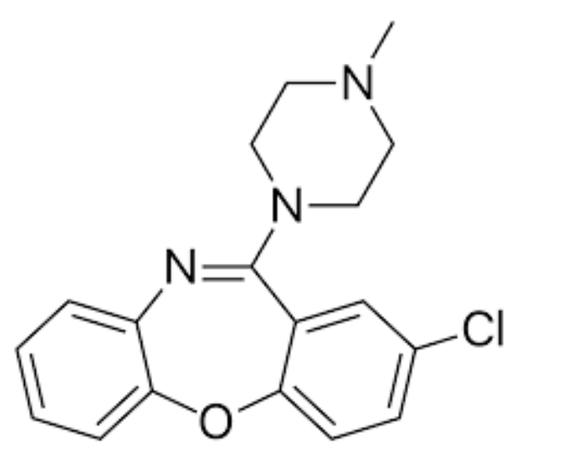

Kim et al, Nature Communications, 2021

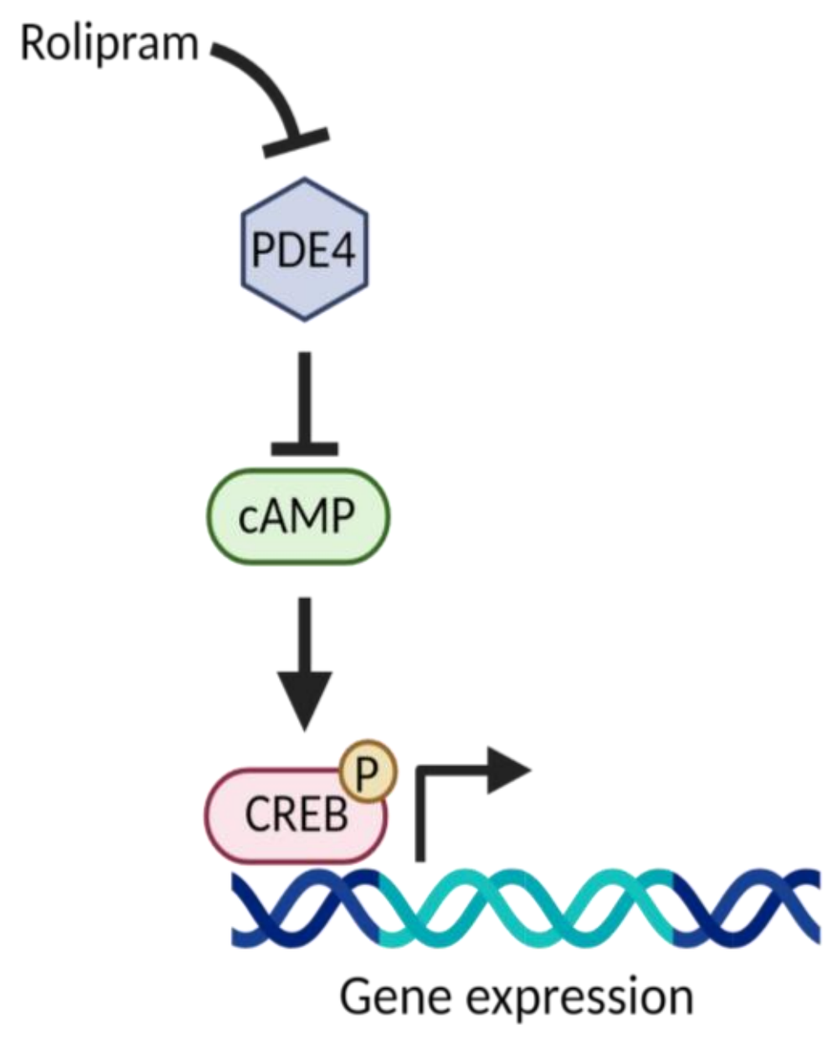

Rolipram
PDE4
cAMP
CREB
P
Gene expression

Unsupervised classification

All image-based features

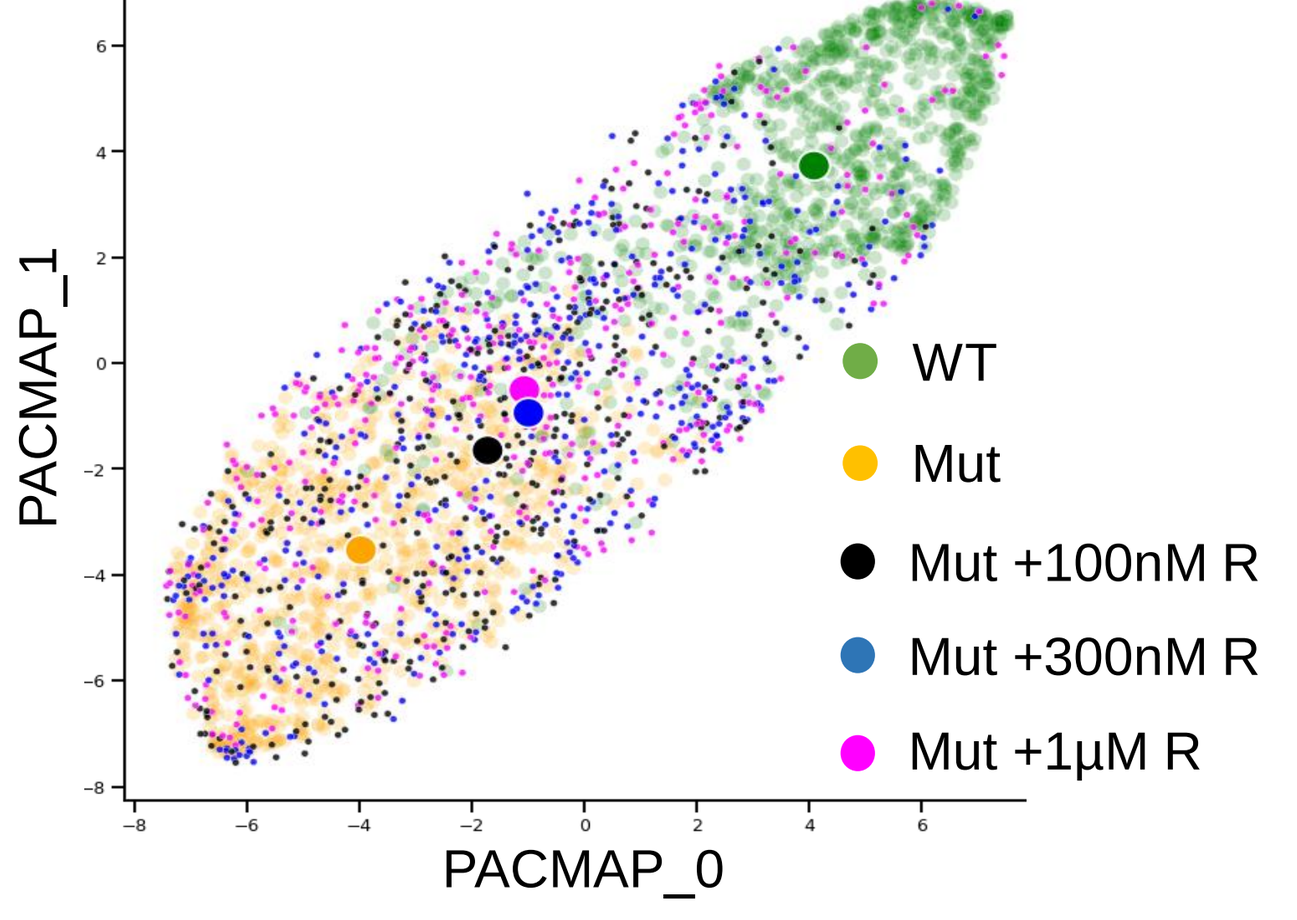

PACMAP_1
PACMAP_0
WT
Mut
Mut +100nM R
Mut +300nM R
Mut +1µM R

Supervised classification (LDA)

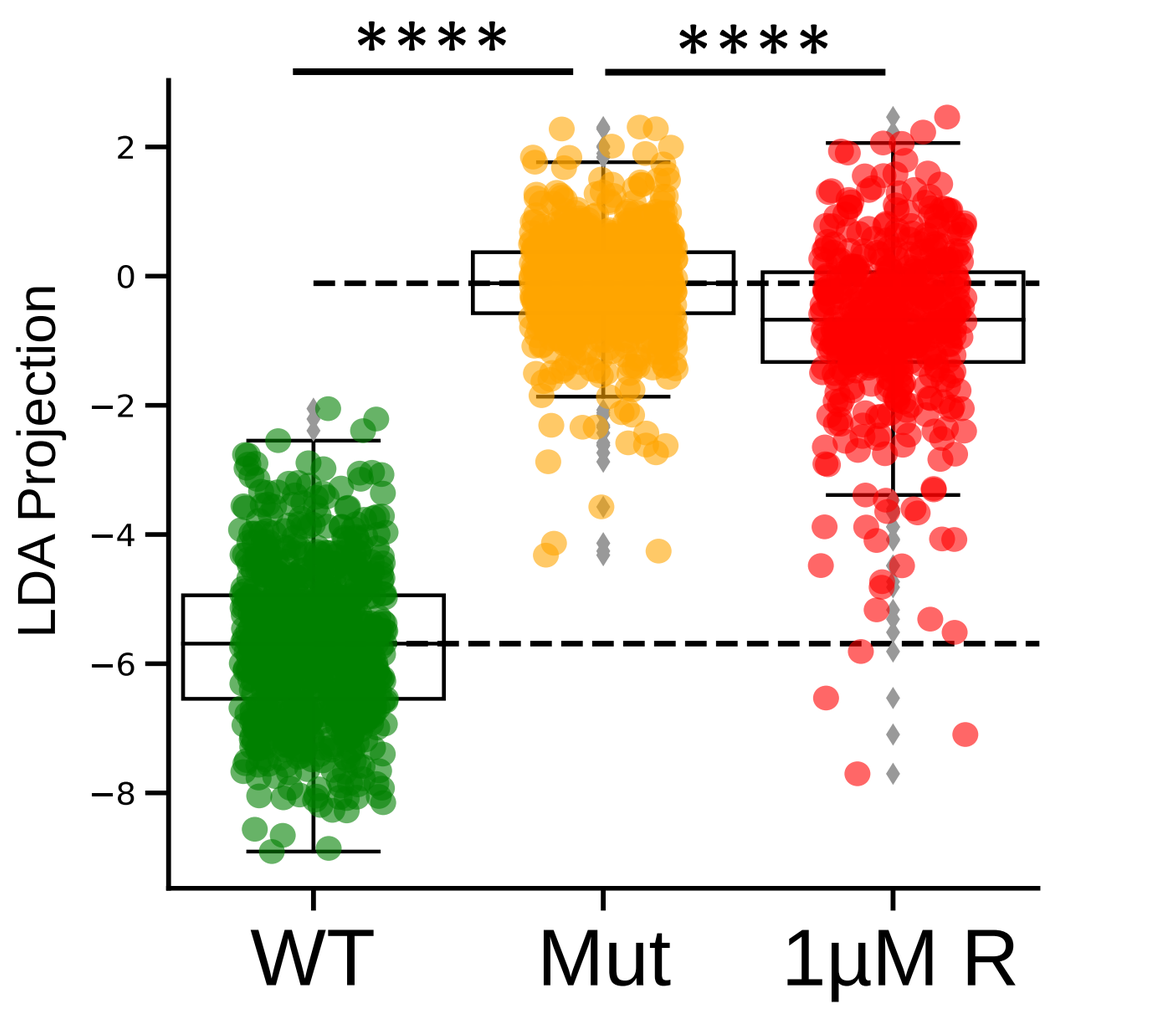

LDA Projection
WT
Mut
1µM R

LDA Rescue

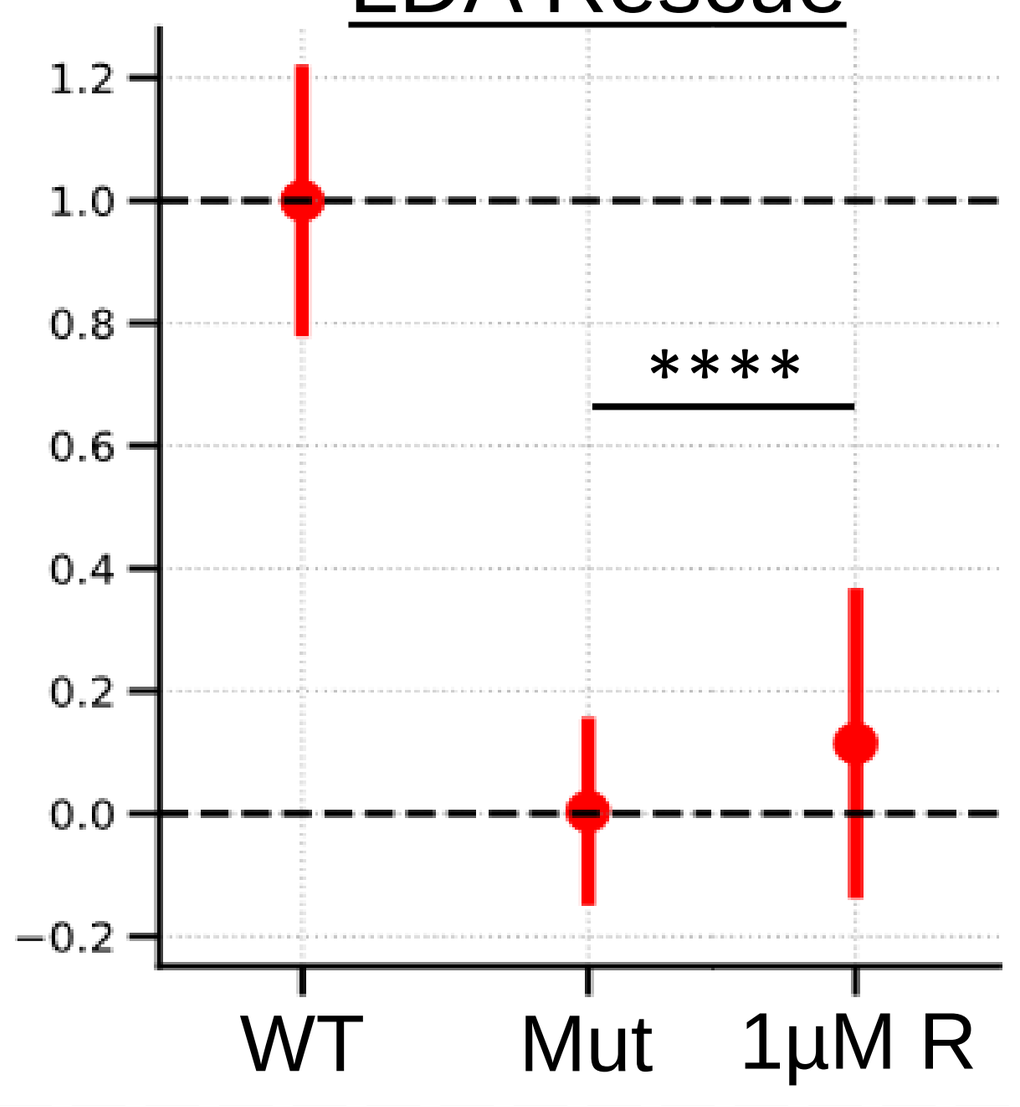

WT
Mut
1µM R

Rolipram fingerprint

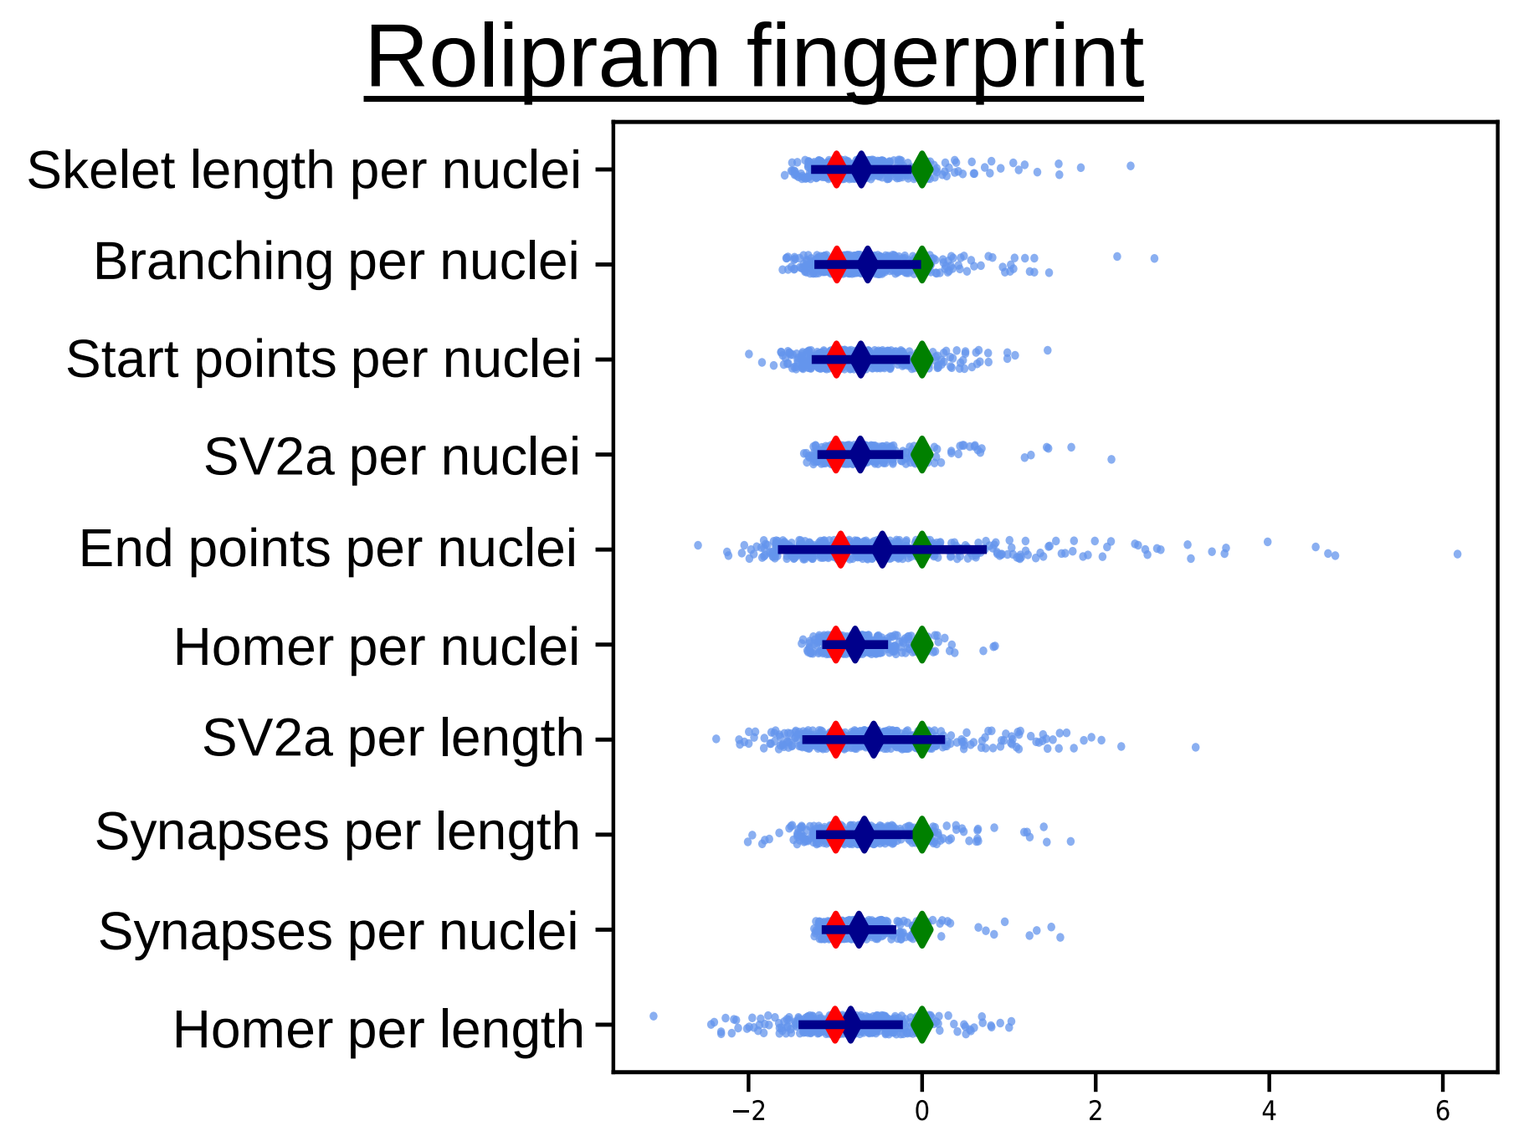

Skelet length per nuclei
Branching per nuclei
Start points per nuclei
SV2a per nuclei
End points per nuclei
Homer per nuclei
SV2a per length
Synapses per length
Synapses per nuclei
Homer per length

Lithium Chloride (Li)

Li-Cl

EU 1794-27 (EU)

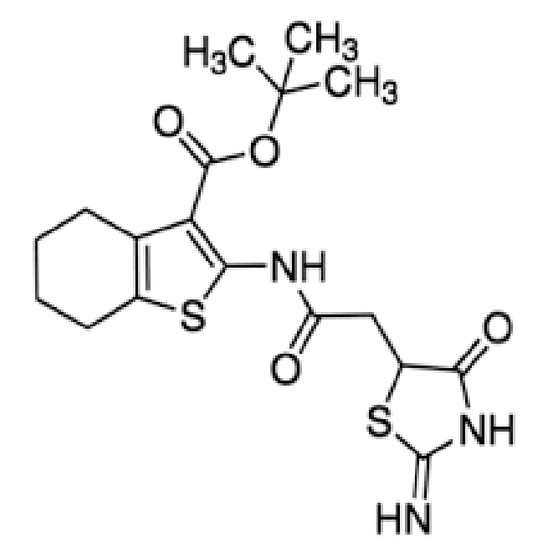

Supervised classification (LDA)

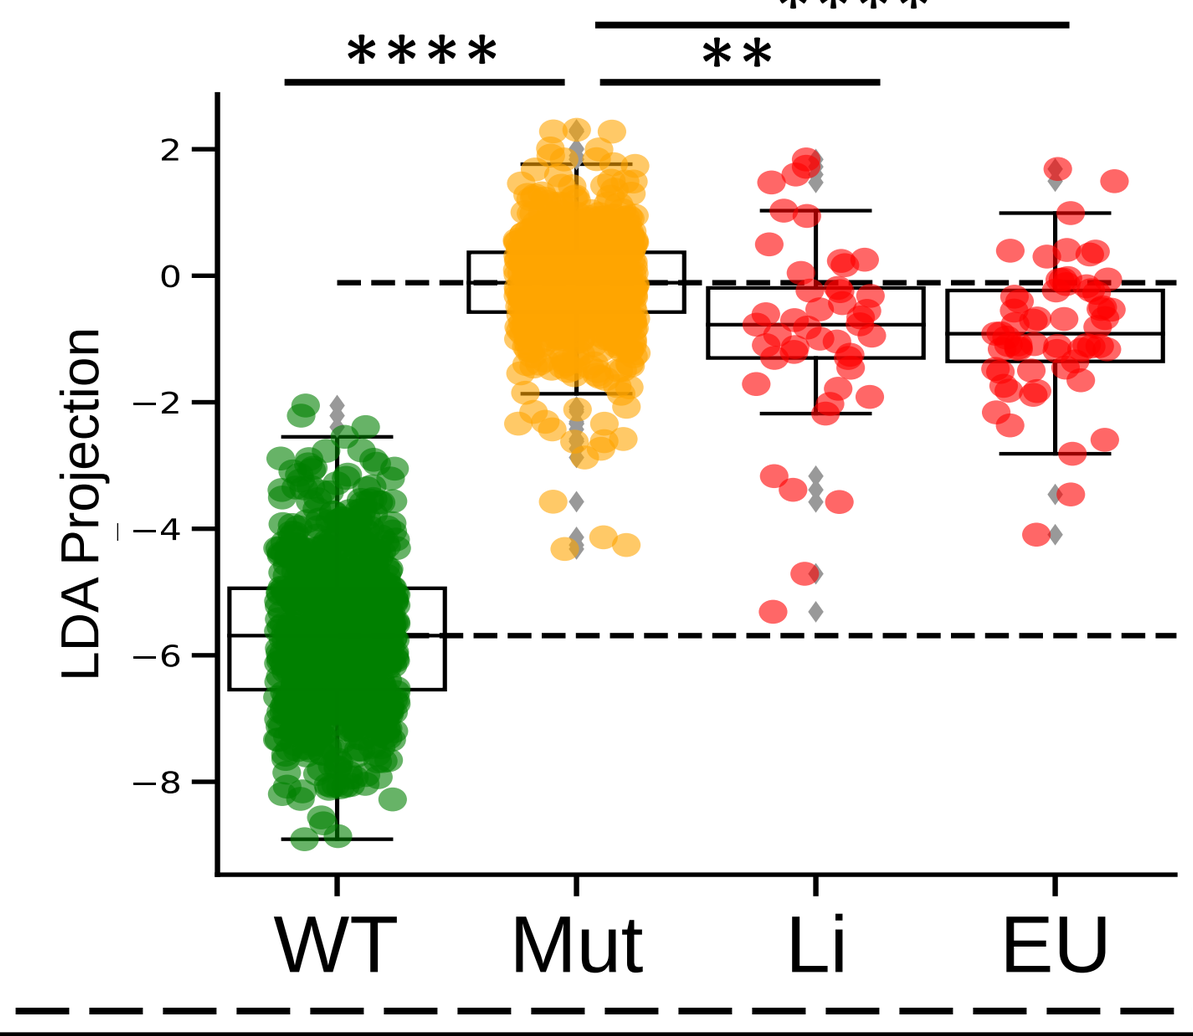

**

LDA Projection
WT
Mut
Li
EU

LDA rescue

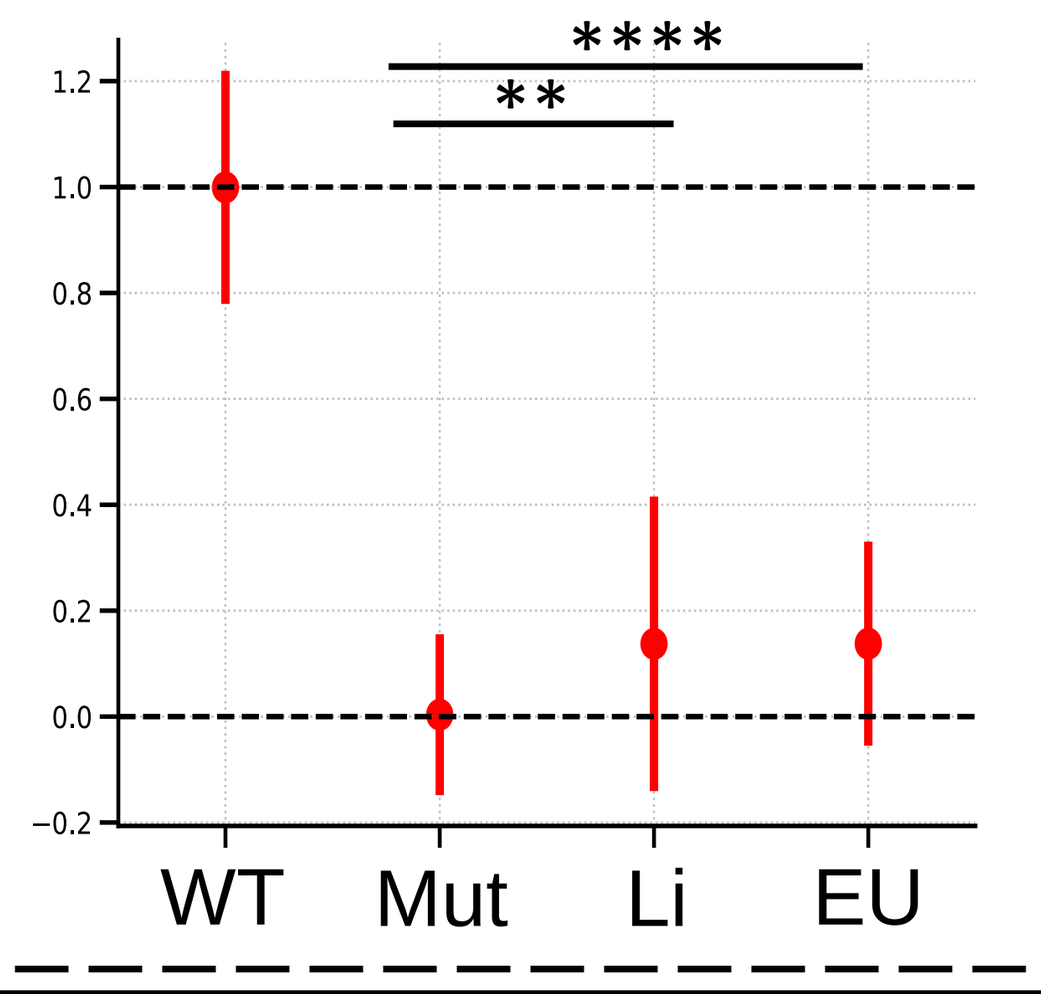

**
WT
Mut
Li
EU

Highlights

- Impaired synaptic phenotypes are recapitulated in DISC1 iPSC-derived neurons
- Assay automation and adaption to 384-well plates
- Machine learning classifies neuronal genotypes based on image-extracted features
- Machine learning identifies drug showing pharmacological rescue in neurons

Contact us for any inquiry on Ksilink:

Karen Schmitt, PhD
Neurobiological Project Team Leader
karen.schmitt@ksilink.com